Pharmacokinetics, Pharmacodynamics, and Safety of Single and Multiple Oral Doses of Vadadustat in Healthy Japanese and Caucasian Subjects

Gurudatt A. Chandorkar,[1] Stanford Jhee,[2] David Han,[3] Kara Schmelzer,[2] Molly Sherman,[2] Karishma Manzur,[1] Amit Sharma,[1] Qing Zuraw,[1] Emil deGoma[1]

[1]Akebia Therapeutics, Inc., Cambridge, MA; [2]PAREXEL International Early Phase, Glendale, CA; [3]California Clinical Trials Medical Group in affiliation with PAREXEL Early Phase, Glendale, CA

Introduction

- Vadadustat (AKB-6548) is an orally dosed, hypoxia-inducible factor prolyl-hydroxylase domain (HIF-PHD) inhibitor currently in Phase 3 trials for the treatment of anemia in patients with non-dialysis-dependent (NDD-) or dialysis-dependent (DD-) chronic kidney disease (CKD)
- In completed Phase 1 and Phase 2 studies, vadadustat demonstrated dose-proportional pharmacokinetics (PK) and pharmacodynamics (PD); and achieved moderate, physiological increases in serum erythropoietin (EPO)[1]
- When administered to patients with NDD-CKD or DD-CKD, vadadustat increased and maintained hemoglobin (Hb) levels in a dose-dependent manner[2–4]
- The ongoing Phase 3 PRO_2TECT and $INNO_2VATE$ programs[5–8] encompass four global, randomized, open-label, cardiovascular outcome trials designed to evaluate the efficacy and safety of vadadustat compared with darbepoetin alfa in patients with NDD-CKD or DD-CKD
- Given the global burden of CKD and the potential for vadadustat to be used for treating anemia secondary to CKD worldwide, this study was conducted to characterize and compare the PK, PD and safety of vadadustat between healthy Japanese and Caucasian subjects

Study Design

- Randomized, double-blinded, placebo-controlled, multiple-ascending dose study to assess the PK, PD, safety and tolerability of oral vadadustat in healthy Japanese and Caucasian adult subjects

Eligibility Criteria

- Age: 20–55 years old
- Healthy adult male or female Japanese[1] and Caucasian[2] subjects
- BMI: 18–30 kg/m²
- Body weight: 45–90 kg (Japanese) or 50–100 kg (Caucasian)

Ethnicity

Japanese (n=24)

Caucasian (n=24)

Test Groups

Daily dosing for 10 consecutive days

Vadadustat 600 mg QD (n=12)

Vadadustat 300 mg QD (n=12)

Vadadustat 150 mg QD (n=12)

Placebo (n=12)

Subjects were randomized 3:1 to receive vadadustat or placebo

Endpoints

These endpoints were measured following single and multiple daily doses:

- PK and PD parameters[3]
- Safety and tolerability

BMI, body mass index; QD, once daily.

1. The following criteria were specific to Japanese subjects: 1) the subject was born in Japan; 2) the subject had 2 biological Japanese parents and 4 Japanese grandparents, as confirmed by interview; 3) the subject had been living outside of Japan for ≤10 years at the time of the screening visit; 4) the subject's lifestyle, including diet, had not changed significantly since leaving Japan.
2. Of European or Latin American descent.
3. Plasma samples were collected over a 24-hour period on Days 1 and 10 and analyzed for vadadustat and metabolites (acyl- and O-glucuronide) by validated LC/MS/MS methods. Standard PK parameters were calculated using non-compartmental methods.

Demographics and Baseline Characteristics

- Demographics and baseline characteristics were generally balanced between Japanese and Caucasian subjects across treatment groups
- All 48 subjects enrolled in the trial completed the study and were included in the PK, PD, and safety analyses

	Vadadustat (all doses)		Placebo	
	Japanese (n=18)	Caucasian (n=18)	Japanese (n=6)	Caucasian (n=6)
Gender, male	13 (72.2)	18 (100.0)	5 (83.3)	6 (100.0)
Age, years	35.7 ± 7.2	35.6 ± 9.0	33.3 ± 6.6	28.3 ± 4.8
Body-Mass Index, kg/m²	22.2 ± 2.7	25.1 ± 2.9	22.4 ± 2.6	23.6 ± 2.5
Weight, kg	64.1 ± 10.5	77.8 ± 25.1	67.6 ± 9.7	73.3 ± 12.0
eGFR, mL/min/1.73 m²	108.1 ± 22.4	115.8 ± 23.1	110.0 ± 18.5	116.3 ± 21.0
Hemoglobin, g/dL	14.9 ± 1.4	15.4 ± 0.8	14.5 ± 0.8	15.5 ± 0.7

Data are presented as n (%) or mean ± standard deviation.

Results

Mean Vadadustat Concentration-Time Profiles Following Multiple Doses (10 Days) of Daily Vadadustat Dosing

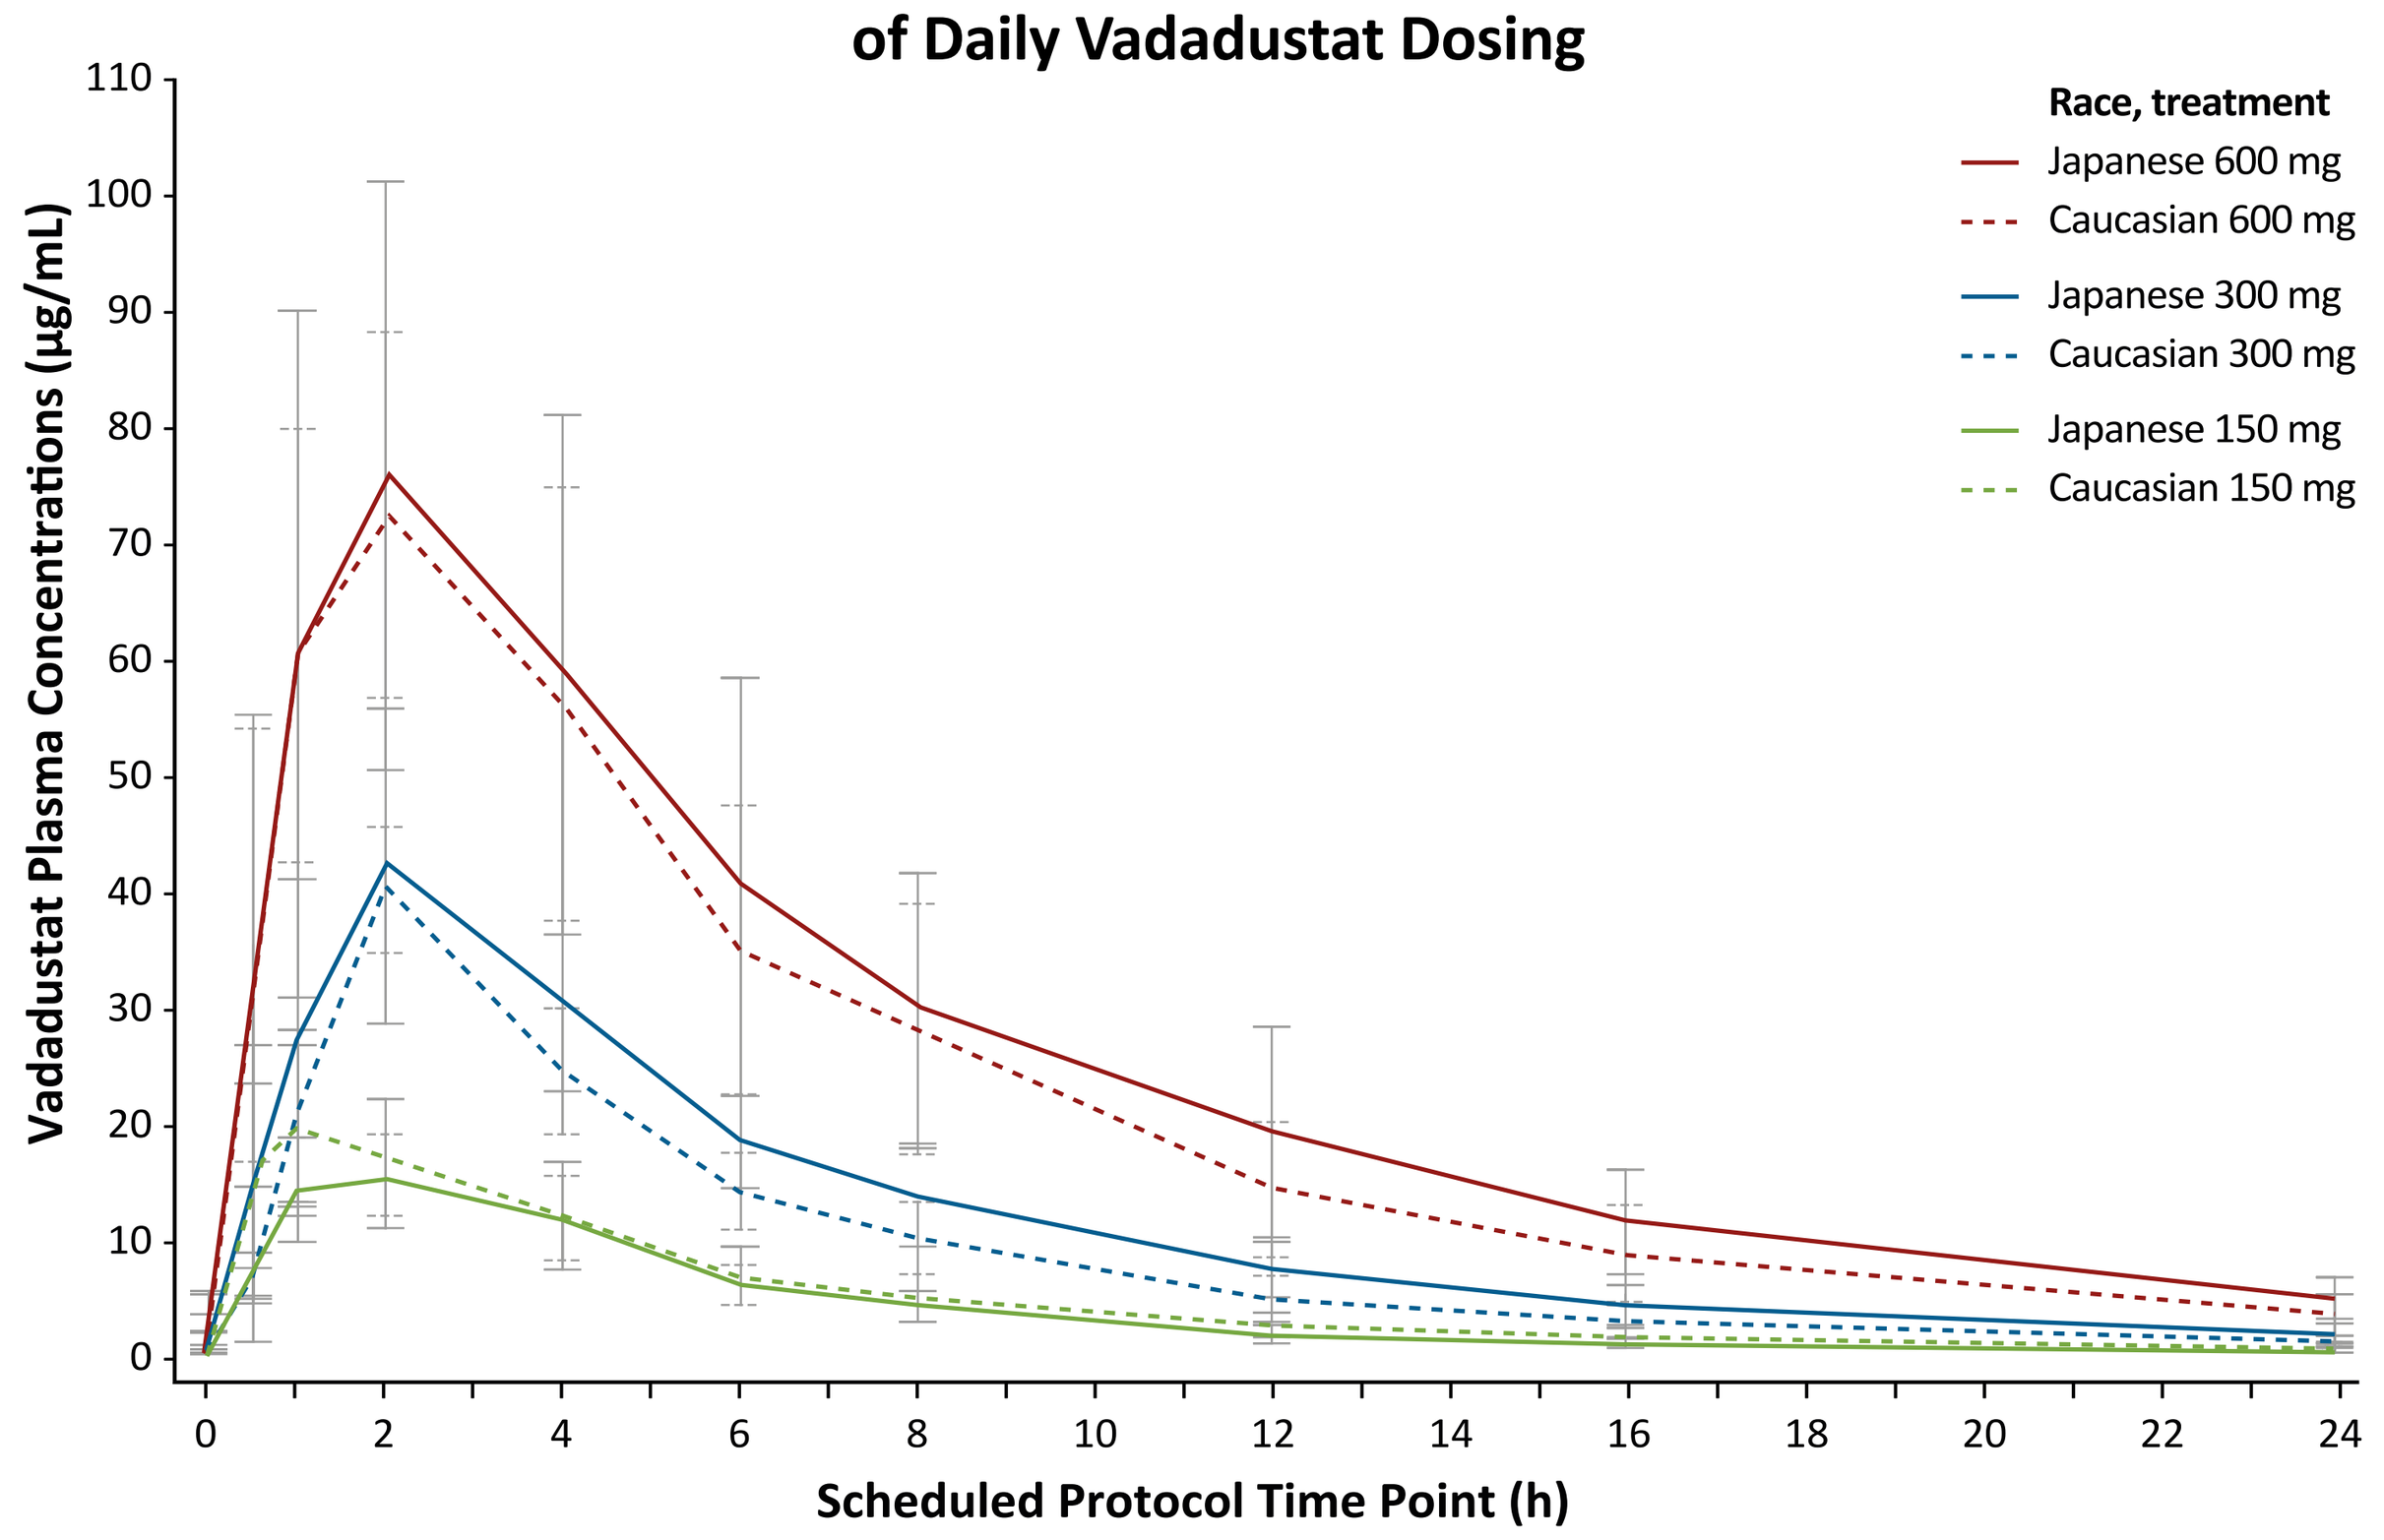
Race, treatment
Japanese 600 mg
Caucasian 600 mg
Japanese 300 mg
Caucasian 300 mg
Japanese 150 mg
Caucasian 150 mg
Vadadustat Plasma Concentrations (µg/mL)
Scheduled Protocol Time Point (h)

Error bars represent standard deviation.

- Similar concentration-time profiles were observed after a single (Day 1) administration of vadadustat

Key PK Parameters Following Multiple Doses (10 Days) of Daily Vadadustat Dosing

	Dose (mg)	n	$AUC_{0-\tau}$ (hr*µg/mL)	$C_{max,ss}$ (µg/mL)	$t_{max,ss}$ (h)	$t_{½,ss}$ (h)	$CL/F_{,ss}$ (L/h)	$Vz/F_{,ss}$ (L)
Japanese	**150**	6	123 ± 30.5	24.2 ± 5.0	0.75 (0.45–3.93)	5.96 ± 0.91	1.29 ± 0.36	10.8 ± 2.09
Japanese	**300**	6	289 ± 75.3	44.3 ± 10.8	1.95 (1.99–4.00)	6.14 ± 0.76	1.1 ± 0.29	9.73 ± 2.53
Japanese	**600**	6	624 ± 205	84.8 ± 22.3	1.98 (0.98–4.00)	6.07 ± 0.42	1.04 ± 0.29	9.16 ± 2.78
Caucasian	**150**	6	102 ± 22.3	18.0 ± 1.7	2.00 (1.00–4.03)	6.05 ± 0.85	1.54 ± 0.37	13.6 ± 3.95
Caucasian	**300**	6	226 ± 47.3	40.4 ± 5.5	2.02 (1.95–2.05)	5.55 ± 0.74	1.39 ± 0.33	11.0 ± 2.48
Caucasian	**600**	6	556 ± 154	79.0 ± 14.4	1.53 (0.97–4.02)	5.64 ± 0.86	1.16 ± 0.36	9.23 ± 2.18

Data are presented as mean ± standard deviation or median (minimum – maximum).
$AUC_{0-\tau}$, area under the concentration-time curve (AUC) over the dosing interval; $CL/F_{,ss}$, apparent clearance at steady-state following oral administration for the parent drug; C_{max}, maximum concentration; t_{max}, time of maximum plasma concentration; $t_{½,ss}$, terminal elimination half-life at steady state (Day 10); $V_z/F_{,ss}$, apparent volume of distribution during terminal phase at steady-state for the parent drug.

- Similar PK parameters were observed after a single day (Day 1) administration of vadadustat

Safety

- Mild adverse events (AEs) were reported in one Japanese subject (skin irritation) and one Caucasian subject (dizziness). The events were assessed as possibly related to study drug and resolved by the end of the study without intervention or cessation of vadadustat dosing
- No deaths or serious AEs were reported during the study

Results (continued)

Mean EPO Concentration-Time Profiles Following Daily Vadadustat Administration

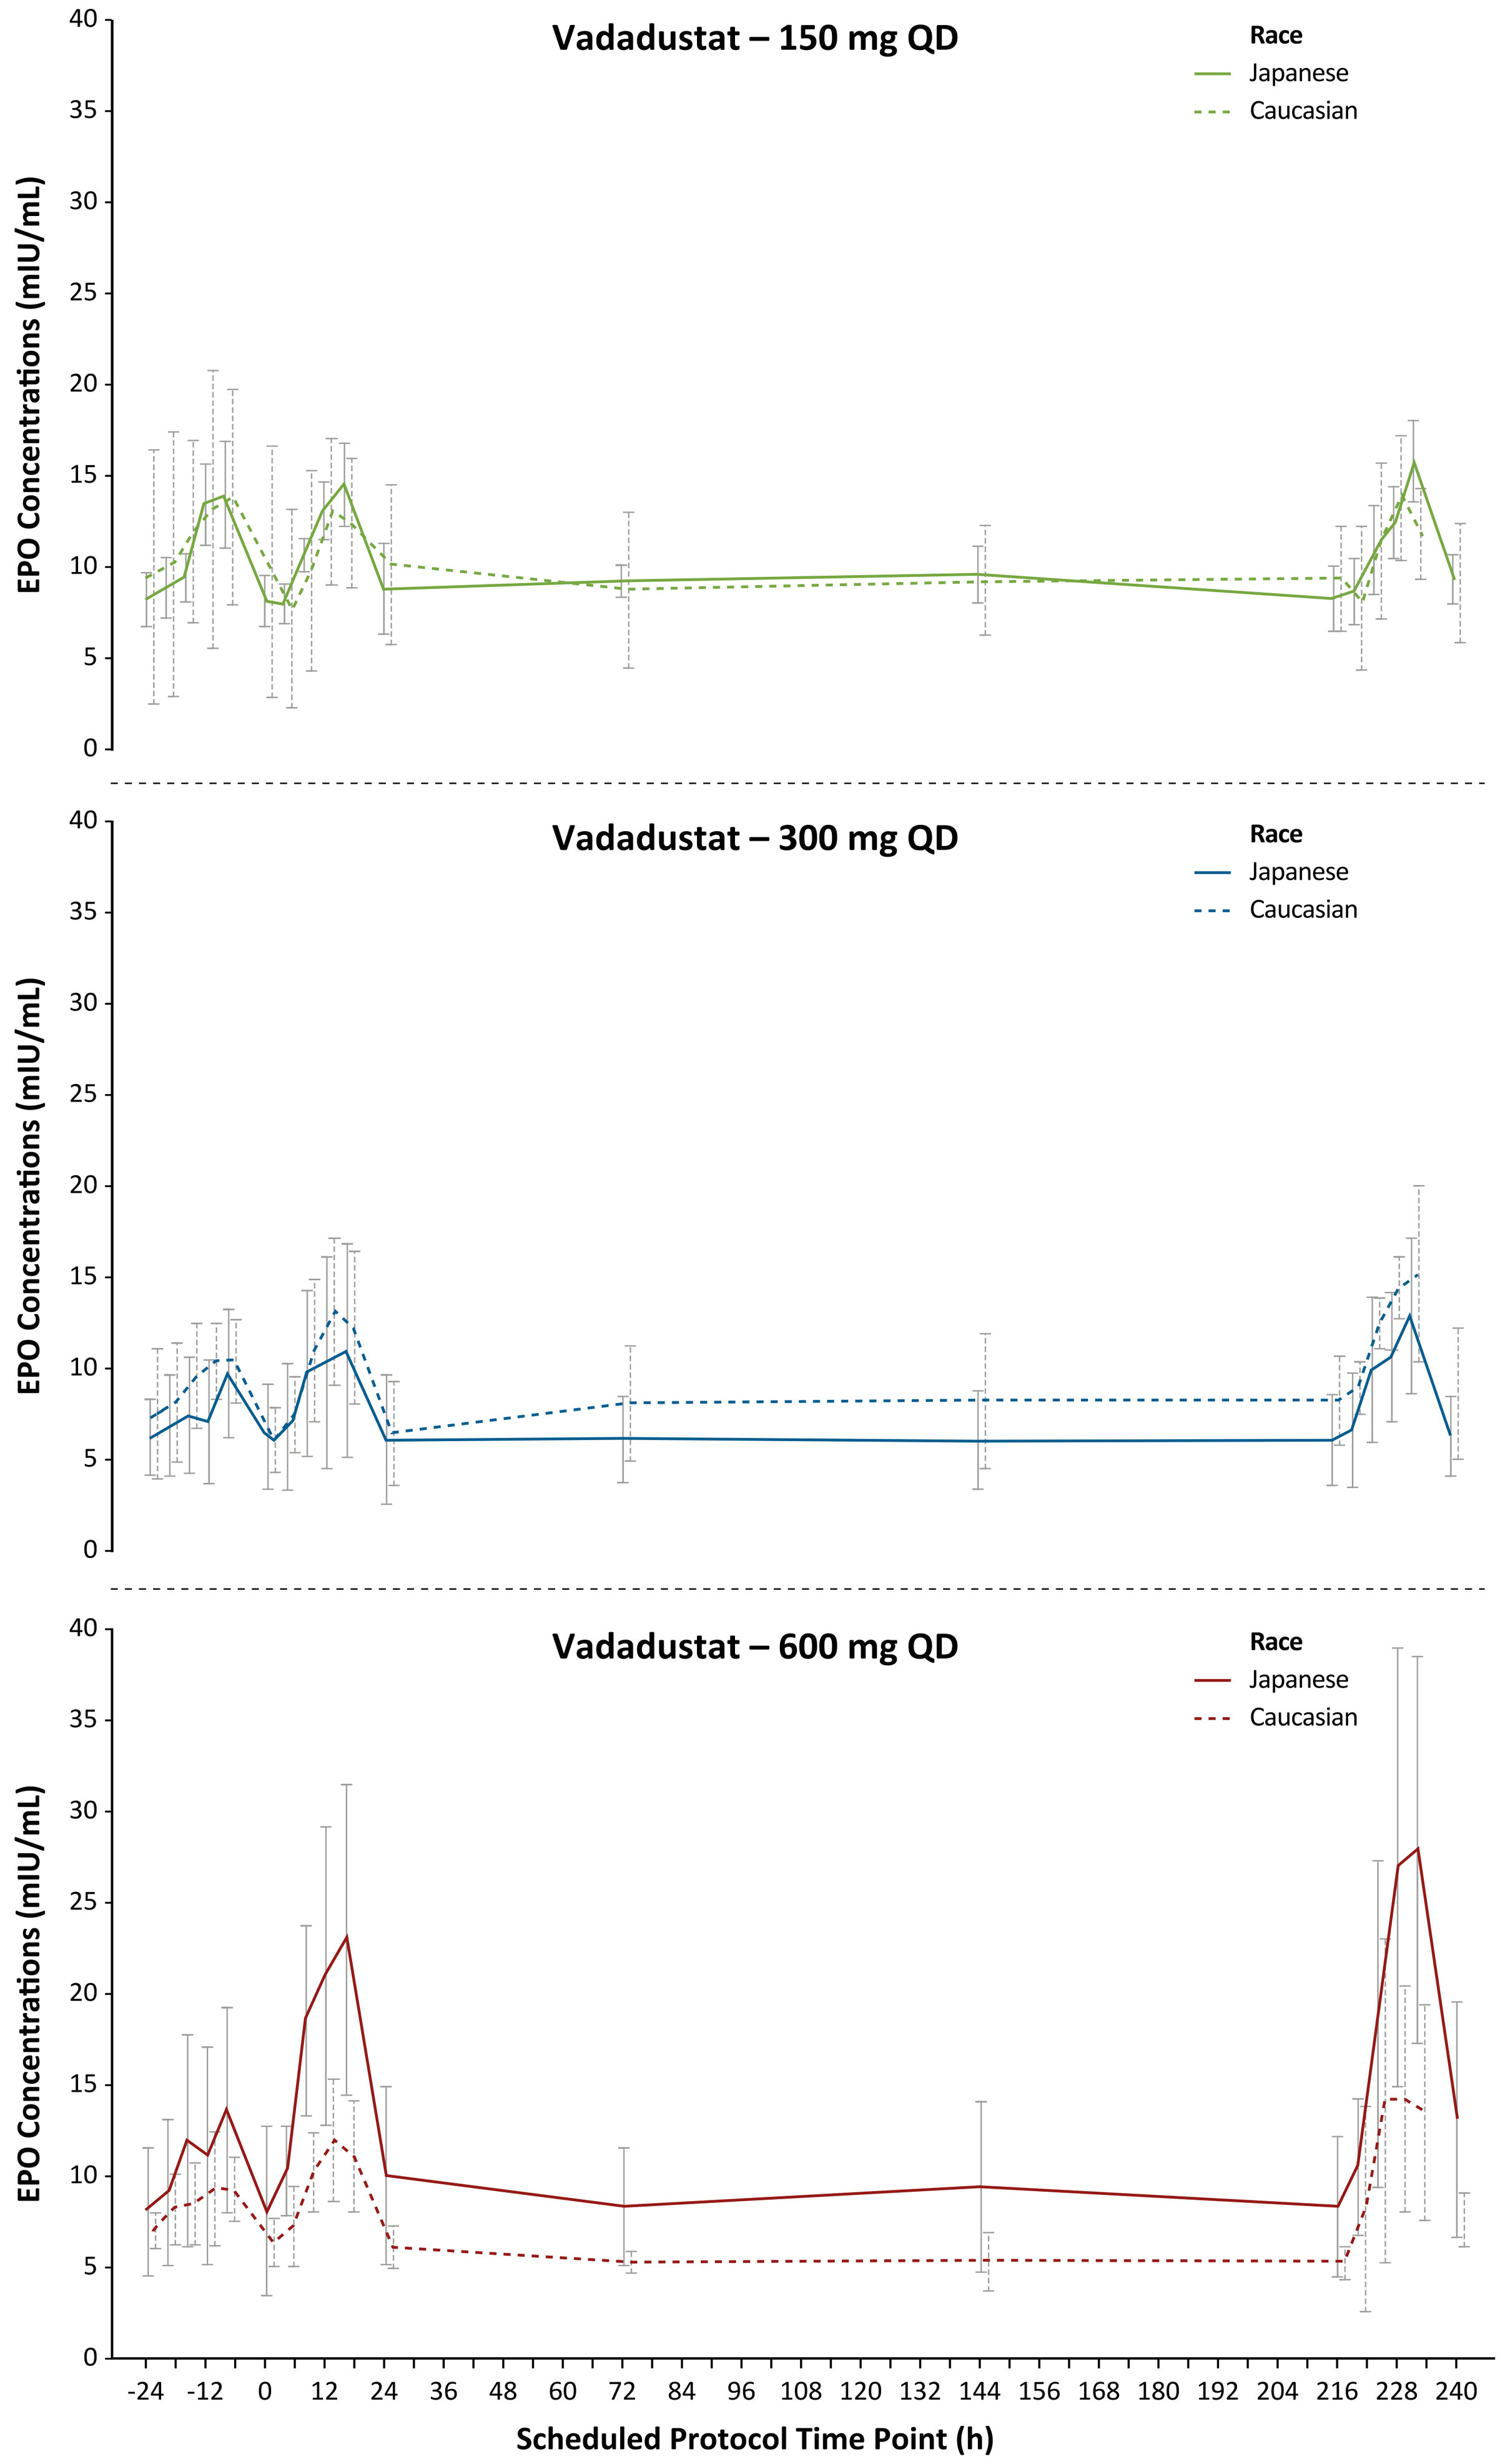
Vadadustat – 150 mg QD
Vadadustat – 300 mg QD
Vadadustat – 600 mg QD
Race
Japanese
Caucasian
EPO Concentrations (mIU/mL)
Scheduled Protocol Time Point (h)

Error bars represent standard deviation.

Results (continued)

Mean EPO Concentration-Time Profiles Following Daily Vadadustat Administration

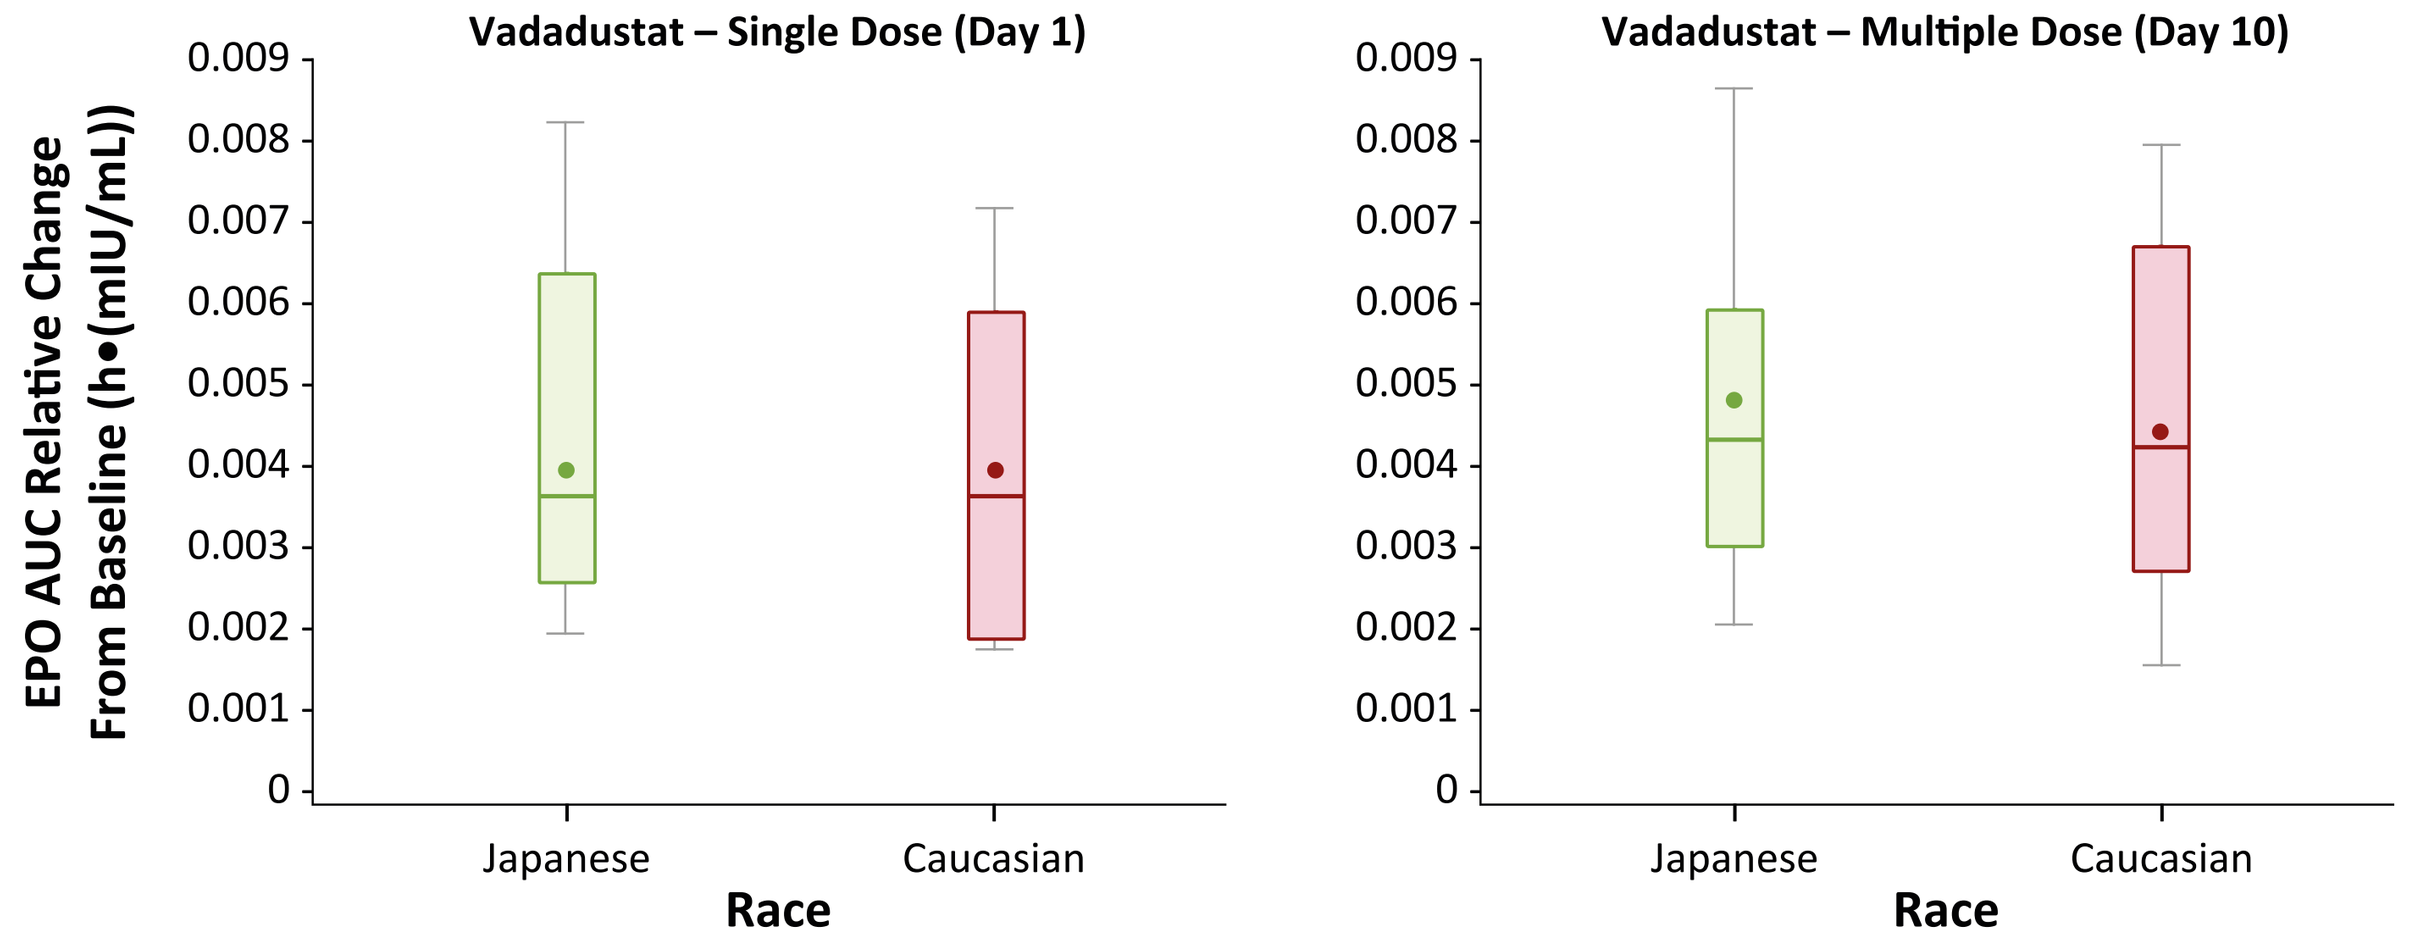
Vadadustat – Single Dose (Day 1)
Vadadustat – Multiple Dose (Day 10)
EPO AUC Relative Change From Baseline (h•(mIU/mL))
Japanese
Caucasian
Race

IQR, interquartile range.

Box plots represent 25th and 75th percentiles; whiskers represent lowest/highest data values within 1.5 IQR of the lower/upper quartiles; median is the line within the box; mean is the dot within the box.

Conclusions

- This study demonstrated that vadadustat exposure was similar in healthy Japanese and Caucasian subjects administered single and multiple vadadustat daily doses
- Vadadustat administration preserved the normal diurnal pattern of EPO secretion and maintained serum EPO levels within the physiologic range
- Plasma concentrations of the metabolites acyl-glucuronide and O-glucuronide were low compared with vadadustat following single and multiple dose administration and were similar between the two ethnicities
- There were no meaningful differences in serum EPO concentrations in healthy Japanese and Caucasian subjects administered single and multiple vadadustat daily doses
- Vadadustat was well-tolerated in healthy Japanese and Caucasian adults
- The similarities in exposure to vadadustat and serum EPO between Japanese and Caucasian subjects support continued development of vadadustat in Japan

PK/PD Summary

- Vadadustat exposure in Japanese and Caucasian subjects increased in a dose-proportional manner across dose levels following single and multiple doses of 150, 300, and 600 mg once daily
- Across both ethnicities, the O-glucuronide-to-parent metabolic ratio (MR) was low (7–10%) across vadadustat dose levels following single and multiple daily doses; the acyl-glucuronide MR could not be calculated due to low concentrations
- Administration of vadadustat preserved the normal diurnal pattern of EPO secretion and maintained serum EPO levels within the physiologic range
- No effect of ethnicity was observed on serum EPO levels following administration of 150 and 300 mg single and multiple doses of vadadustat
- Serum EPO concentrations in Japanese subjects at the 600 mg dose were slightly higher than in Caucasian subjects, but these slight differences were not considered clinically significant

References

1. Buch A, Maroni B, Hartman C. *J Am Soc Nephrol* 2015;26:747A.
2. Pergola PE, Spinowitz BS, Hartman CS, et al. *Kidney Int* 2016;90:1115–22.
3. Haase VH, Hartman CS, Maroni B, et al. *National Kidney Foundation Spring Clinical Meeting 2016.* Abstract 171.
4. Hartman CS, Shalwitz IR, Shalwitz RA. *Nephrol Dial Transplant* 2014;29(Supplement 3): II22, Abstract SO051.
5. NCT02892149. Available at: clinicaltrials.gov. Last accessed: November 2016.
6. NCT02680574. Available at: clinicaltrials.gov. Last accessed: November 2016.
7. NCT02865850. Available at: clinicaltrials.gov. Last accessed: November 2016.
8. NCT02648347. Available at: clinicaltrials.gov. Last accessed: November 2016

Acknowledgments

The authors thank Dr. Ramin Farzaneh-Far for initial participation in abstract development for this poster, and Wenli Luo for help with statistical analysis. We also thank Lucid Partners Ltd. for poster layout.
The study was funded by Akebia Therapeutics, Inc. To download an electronic copy of the poster, please visit: **www.akebia.com/media/publications.**

Akebia™ THERAPEUTICS